Ivospemin and doxorubicin combination modulates polyamine metabolism to improve survival in a murine ovarian cancer model

Cassandra E. Holbert[1], Jackson R. Foley[1], Ashley C. Nwafor[1], Ting-Ann Liu[1], Elizabeth Bruckheimer[2], Jennifer K. Simpson[2], Tracy Murray Stewart[1], Robert A. Casero, Jr.[1]

[1]Johns Hopkins Medicine, Sidney Kimmel Comprehensive Cancer Center, Baltimore MD

[2]Panbela Therapeutics, Waconia MN

Abstract #7154

Polyamines are small polycationic alkylamines that are absolutely required for the continual growth and proliferation of cancer cells. As cancer cells maintain elevated polyamine pools through dysregulated polyamine metabolism, its pharmacological modulation is a promising avenue in cancer therapeutics. The polyamine analogue ivospemin has shown efficacy in slowing pancreatic and ovarian tumor progression in vitro and in vivo and has demonstrated encouraging results in pancreatic cancer clinical trials. Considering nearly 75% of late-stage ovarian cancer patients develop resistance to platinum-based chemotherapies, limiting treatment options, the aim of our study is to determine the potential efficacy of ivospemin in combination with doxorubicin, a common chemotherapeutic used in platinum-resistant ovarian tumors.

We have previously shown that ivospemin exposure decreases polyamine content in a variety of cancer cell lines through downregulation of the polyamine biosynthetic enzyme ornithine decarboxylase (ODC) and induction of the polyamine catabolic enzyme spermidine/spermine-N1-acetyltransferase (SSAT). Here we examine the potential of combining ivospemin with doxorubicin. Ivospemin treatment reduces cell viability in ovarian adenocarcinoma cell lines and increases the toxicity of doxorubicin regardless of cisplatin sensitivity. Cells treated with the combination exhibit a greater decrease in polyamine levels than cells treated with either single agent. This increased polyamine depletion and decreased survival is accomplished through modulation of polyamine metabolism, predominately through an additive induction of SSAT activity.

Using the syngeneic VDID8+ ovarian murine model, we further evaluated the ability of ivospemin to improve response to doxorubicin at clinical and sub-clinical dosing. Ascites fluid was used as a marker for tumor burden and evaluated for polyamine content. We found that the combination treatment increases median survival, delays tumor onset, and decreases overall tumor burden compared to either clinical or subclinical doxorubicin dosing schemes. Combination treatment also decreases overall polyamine content in the ascites by 75%. N1-acetylated spermidine is enriched in ascites from combination-treated mice, consistent with an upregulation of SSAT in response to treatment. Recognizing the non-representative mutational status of the VDID8+ model as a limitation, we are currently evaluating the combination of ivospemin and doxorubicin in genetically defined murine models that better recapitulate human high-grade serous ovarian carcinomas. Ongoing studies will determine influences on the tumor microenvironment and will mechanistically evaluate the cooperativity of ivospemin and doxorubicin on pathways outside of polyamine metabolism.

Ivospemin

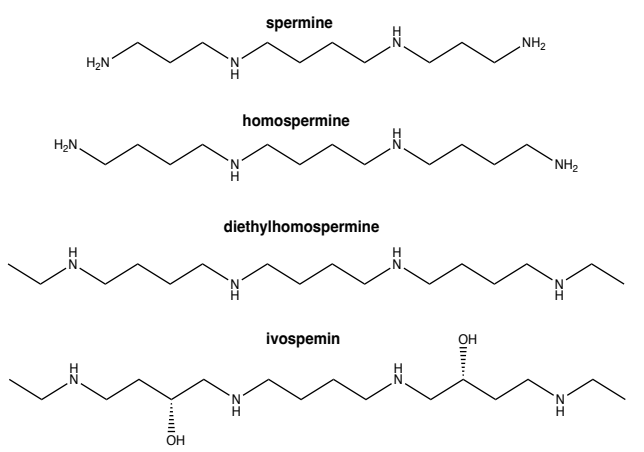

spermine
homospermine
diethylhomospermine
ivospemin

Figure 1. Chemical structures of spermine analogues and their derivatives, including ivospemin. Homospermine is an analogue of spermine produced by the symmetrical addition of two methylene groups. It is further derived to diethylhomospermine by the addition of ethyl groups to either end of the molecule. Ivospemin is a hydroxylated derivative of diethylhomospermine currently under phase 2/3 evaluation in pancreatic cancer.

Results

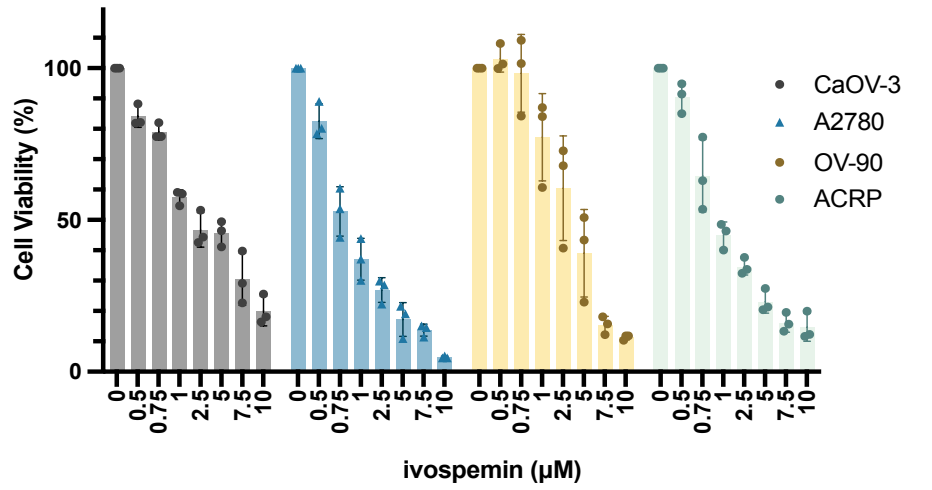

Cell Viability (%)
CaOV-3
A2780
OV-90
ACRP
ivospemin (μM)

Figure 2. Ivospemin treatment reduces cell viability in four human ovarian adenocarcinoma cell lines *in vitro* regardless of cisplatin sensitivity. Ovarian adenocarcinoma cell lines were treated for 96 hours with increasing concentrations of ivospemin ranging from 500 nM to 10 μM. Ivospemin treatment decreased cell viability in all four cell lines tested. These four lines represent varying levels of cisplatin sensitivity with CaOV-3 being the most sensitive and ACRP being the least sensitive.

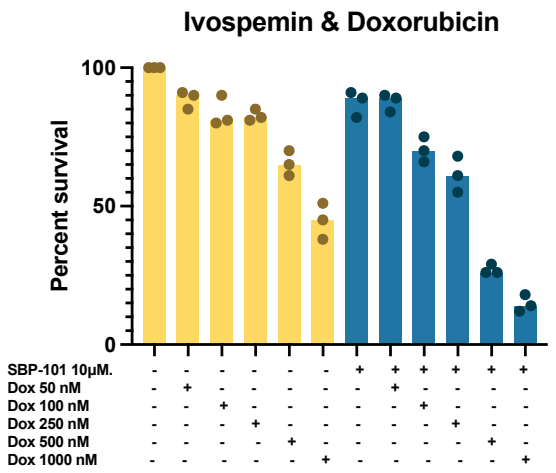

Ivospemin & Doxorubicin
Percent survival
SBP-101 10μM.
Dox 50 nM
Dox 100 nM
Dox 250 nM
Dox 500 nM
Dox 1000 nM

Figure 3. Ivospemin/doxorubicin co-treatment cooperatively reduce cell viability in human ovarian adenocarcinoma cells *in vitro.* The ACRP ovarian adenocarcinoma cell line was treated for 48 hours with increasing concentrations of doxorubicin ranging from 50 nM to 1 μM either with or without 10 μM of ivospemin. Doxorubicin reduced cell viability in a dose dependent manner. The addition of ivospemin treatment notably increased the toxicity of doxorubicin.

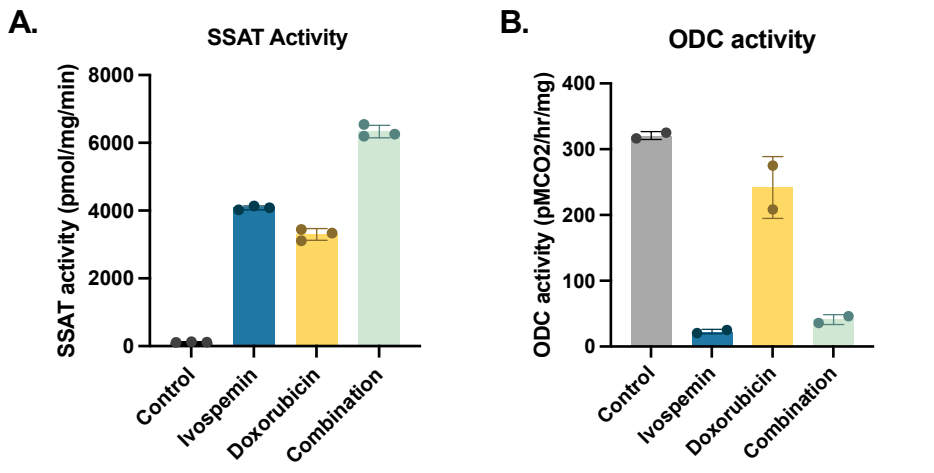

A.
SSAT Activity
SSAT activity (pmol/mg/min)
B.
ODC activity
ODC activity (pMCO2/hr/mg)
Control
Ivospemin
Doxorubicin
Combination

Figure 4. Ivospemin and doxorubicin treatment additively increases polyamine catabolism in human ovarian adenocarcinoma cells *in vitro.* The ACRP ovarian adenocarcinoma cell line was treated for 48 hours with 10 μM of ivospemin and 1 μM doxorubicin. Both agents alone increased the activity of the polyamine catabolic enzyme SSAT with the combination having the highest activity (**A**). While ivospemin decreases polyamine biosynthesis through ODC inhibition, there does not appear to be any additional inhibition with doxorubicin treatment (**B**).

Results

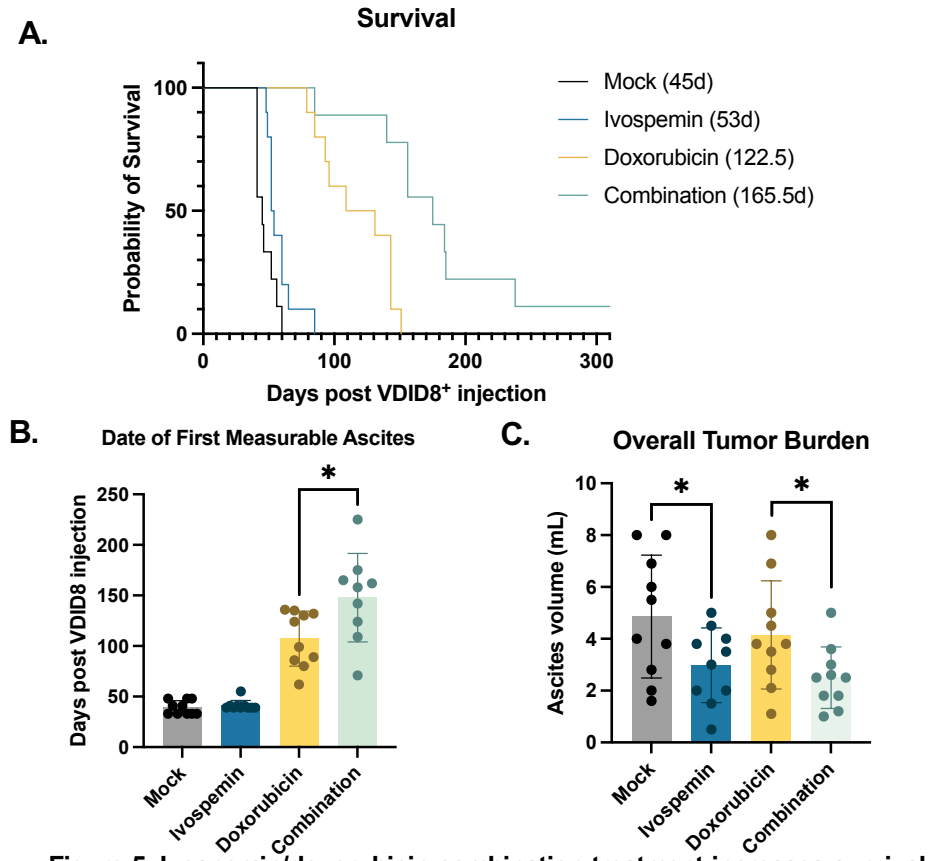

A.
Survival
Probability of Survival
Mock (45d)
Ivospemin (53d)
Doxorubicin (122.5)
Combination (165.5d)
Days post VDID8+ injection
B.
Date of First Measurable Ascites
Days post VDID8 injection
C.
Overall Tumor Burden
Ascites volume (mL)
Mock
Ivospemin
Doxorubicin
Combination

Figure 5. Ivospemin/doxorubicin combination treatment increases survival, delays tumor onset and decreases tumor burden of mice injected with VDID8+ ovarian cancer cells. Utilizing the VDID8+ murine ovarian cancer model (ID8+ C57Bl/6ovarian cells overexpressing both VEGF and Defensin) we treated mice with ivospemin at 24 mg/kg 2qw alt weeks and doxorubicin at 1 mg/kg 3qwx4. Combination treatment increased overall survival by approximately 270% (**A**) which was correlated with an increase in time to ascites production (**B**). Ivospemin monotherapy and combination mice also benefited from a lower overall tumor burden as measured by ascites volume (**C**).

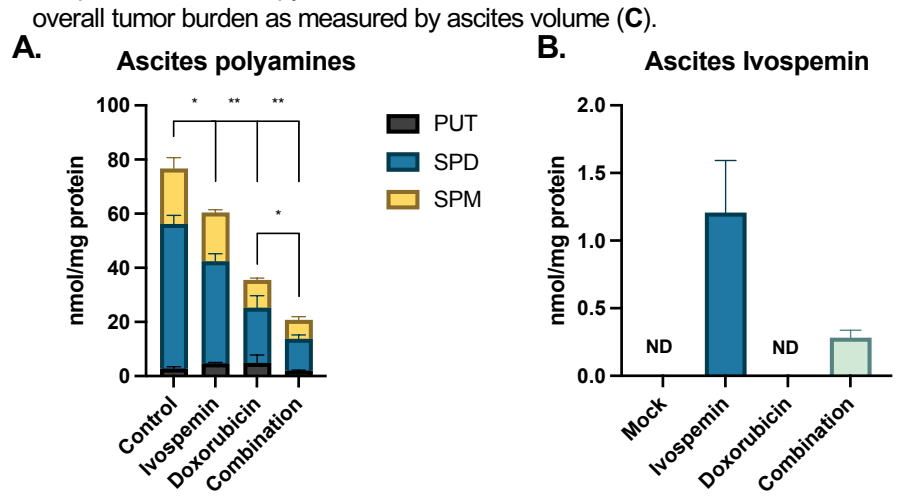

A.
Ascites polyamines
nmol/mg protein
PUT
SPD
SPM
Control
Ivospemin
Doxorubicin
Combination
B.
Ascites Ivospemin
ND
Mock
Ivospemin
Doxorubicin
Combination

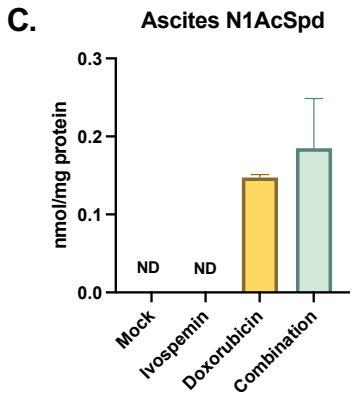

C.
Ascites N1AcSpd
nmol/mg protein
ND
Mock
Ivospemin
Doxorubicin
Combination

Figure 6. Ivospemin and doxorubicin combination treatment decreases overall ascitic polyamine levels. Ascites fluid from treated mice was analyzed by HPLC for polyamine quantification. Both single agents decreased polyamine content with the combination showing the largest decrease (**A**). Ivospemin was successfully quantified in the ascites of only mice exposed to the agent (**B**). Doxorubicin treatment increased the levels of N1-acetylated spermidine suggesting treatment may influence polyamine transport, potentially explaining the slight decrease in ivospemin accumulation in combination mice **(C)**.

Results

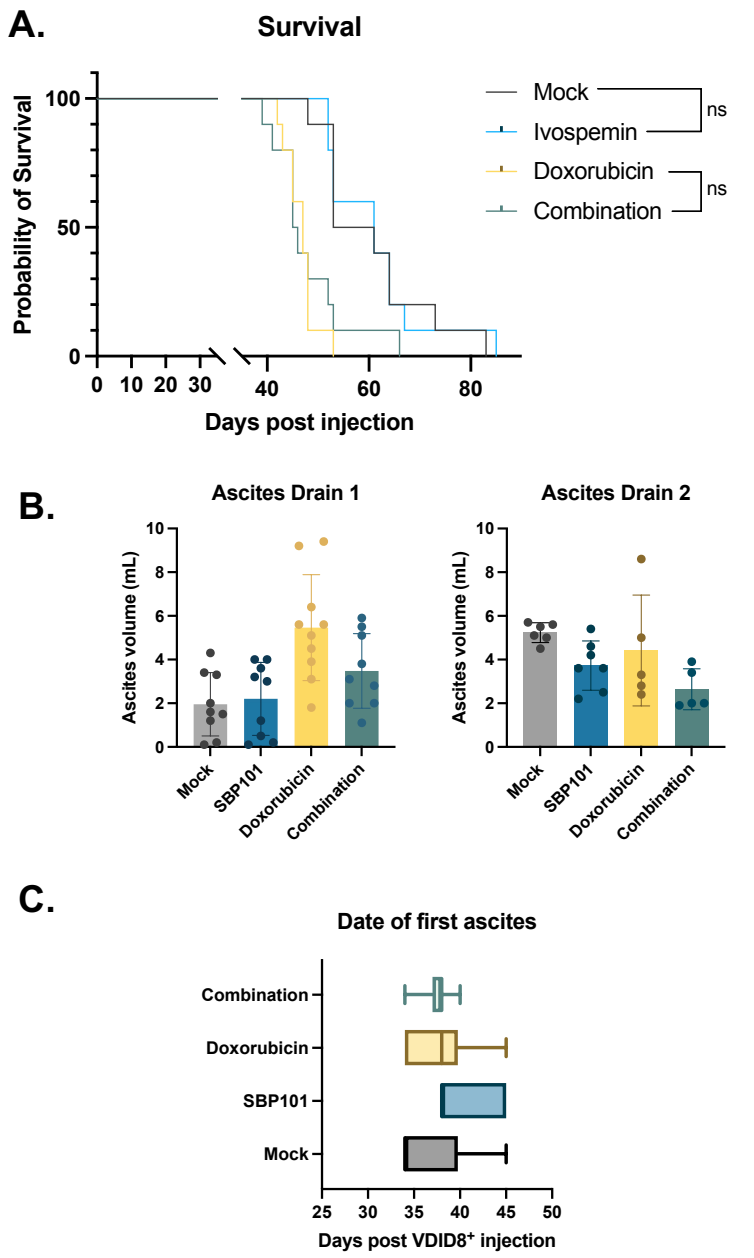

A.
Survival
Probability of Survival
Mock
Ivospemin
Doxorubicin
Combination
ns
Days post injection
B.
Ascites Drain 1
Ascites Drain 2
Ascites volume (mL)
Mock
SBP101
Doxorubicin
Combination
C.
Date of first ascites
Days post VDID8+ injection

Figure 7. Efficacy of the combination treatment is dependent on an intact immune system. Immunocompromised NSG mice were injected with 250,000 VDID8+ cells per mouse and subsequently treated with 24 mg/kg 2qw alt weeks ivospemin (SBP-101) and a subclinical dose of doxorubicin (0.5 mg/kg 2wqx4). Even at a subclinical dosing level, doxorubicin alone produced some toxicities among treated animals, most notably weight loss and anemia, however none of the three treatment arms resulted in a survival benefit **(A)** nor did treatment decrease tumor burden or delay ascites formation as previously seen **(B,C).**

Conclusions

The polyamine analogue, ivospemin, reduces the viability of human ovarian adenocarcinoma cell lines regardless of their platinum sensitivity. Ivospemin and doxorubicin cooperatively increase polyamine catabolism and decrease overall cell survival *in vitro*. The combination significantly prolongs the survival of an immunosuppressive ovarian murine model *in vivo.* Cotreatment also results in delayed ascites formation and decreased overall tumor burden. The combination treatment cooperatively decreases overall ascitic polyamine content. Immunodeficient NSG mice injected with VDID8+ ovarian cancer cells do not receive a survival benefit from ivospemin, doxorubicin, or a combination treatment, indicating that an intact immune system is required for the efficacy of this therapy. Subsequent experiments will aim to determine immune cell populations required for therapeutic efficacy.

PhDPosters.com Your Research Poster, Printed For Less